A route of administration study of BBP-812, an AAV9-based gene therapy for the treatment of Canavan disease, in juvenile cynomolgus macaques

Scott DW, Rouse JL, Romero KB, Eclov R, Lewis TEW, Kapadia M, Mansfield G, and Beard CW
Aspa Therapeutics, BridgeBio, Raleigh, NC , USA

Abstract

Canavan Disease (CD) is a rare pediatric leukodystrophy caused by aspartoacylase deficiency. The disease is characterized by elevated levels of the aspartoacylase substrate N-acetylaspartic acid. Patients present with a lack of psychomotor development and most die by the age of ten. We are developing BBP-812, an AAV9-based gene therapy containing a codon optimized human *Aspa* transgene, to introduce functional aspartoacylase into CD patients. To understand the optimal dosing route for BBP-812 we conducted a study in juvenile cynomolgus macaques comparing: intravenous infusion (IV: doses of 1.8×10^{13}, 6×10^{13}, and 1.8×10^{14} vg/kg), intrathecal lumbar injection (5×10^{12} vg total), or unilateral intracerebroventricular injection (5×10^{12} vg total). Animals were sacrificed three and eight weeks after dosing. During the in-life phase, animals were monitored for changes in hematology and both serum and urine chemistries. At necropsy, biodistribution was measured throughout the brain and spinal cord as vector genomes per diploid cell by droplet digital PCR and transgene RNA was assessed by qRT-PCR. There was a transient increase in ALT and AST in the highest dose IV treatment group at day 3 which returned to normal without intervention by day 8 post dosing. All other markers of hematology, as well as serum and urine chemistries were unchanged. Biodistribution analysis demonstrated that all three routes of administration achieved transduction throughout the spinal cord. However, only animals receiving IV delivered BBP-812 showed transduction throughout deep brain structures critical for the treatment of CD. This work supports the continued development of BBP-812 by intravenous infusion for the treatment of CD.

Background

- Canavan disease is characterized by a loss of Aspa expression and a systemic build up of N-acetylasparitic acid (NAA).
- In the presence of elevated NAA, neuronal demyelination occurs.
- There are currently no therapies that treat the underlying cause of Canavan disease.

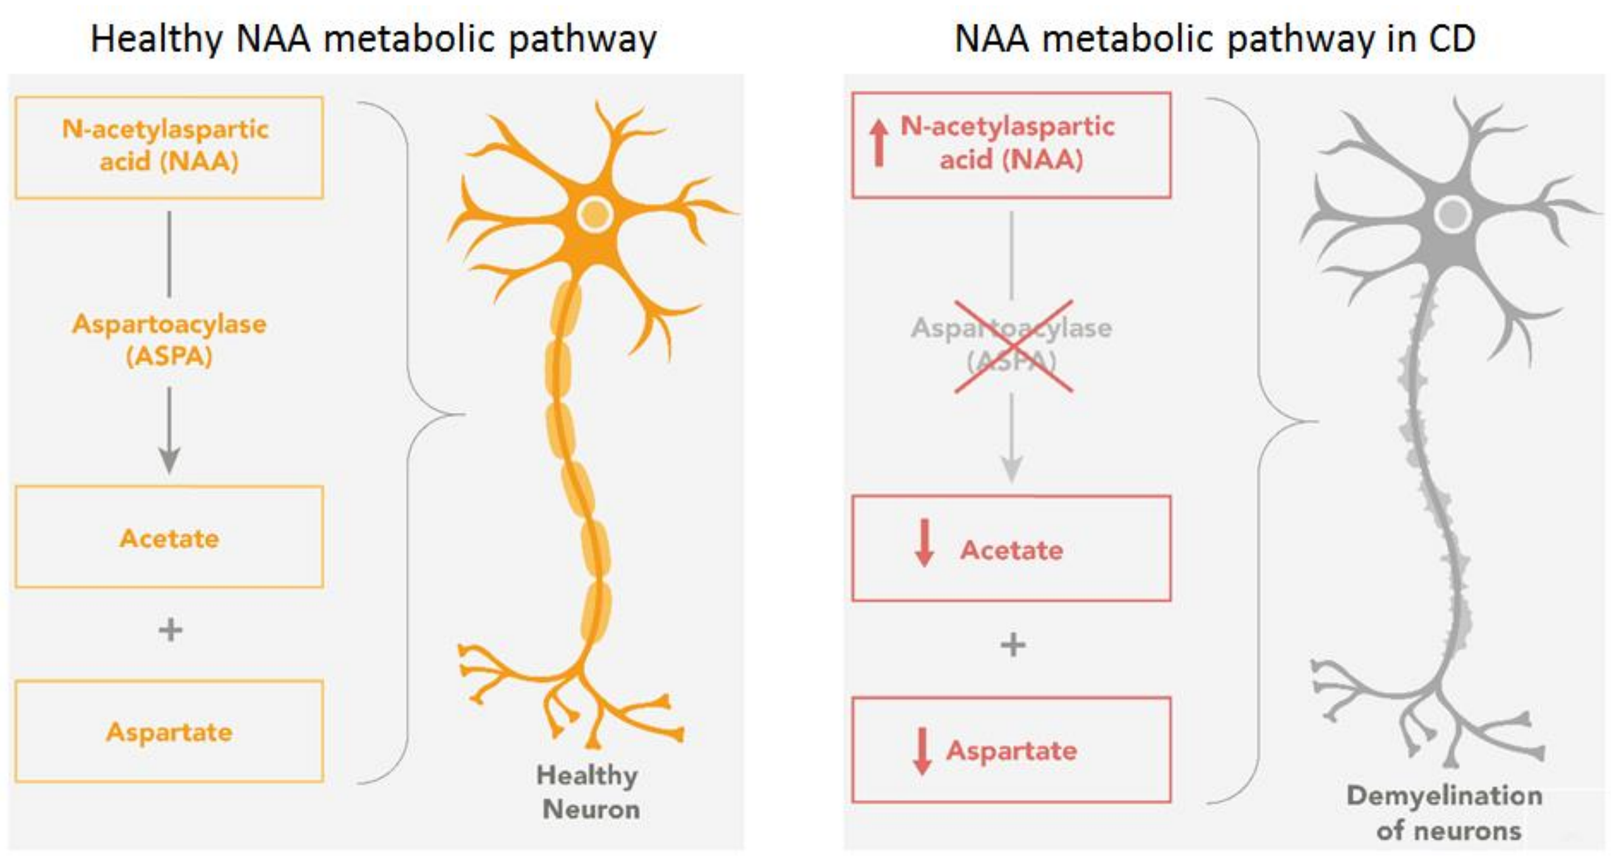
Healthy NAA metabolic pathway
N-acetylaspartic acid (NAA)
Aspartoacylase (ASPA)
Acetate
+
Aspartate
Healthy Neuron
NAA metabolic pathway in CD
N-acetylaspartic acid (NAA)
Aspartoacylase (ASPA)
Acetate
+
Aspartate
Demyelination of neurons

BBP-812 Corrects The Canavan Disease Phenotype In Aspa$^{-/-}$ Mice In A Dose-dependent Manner

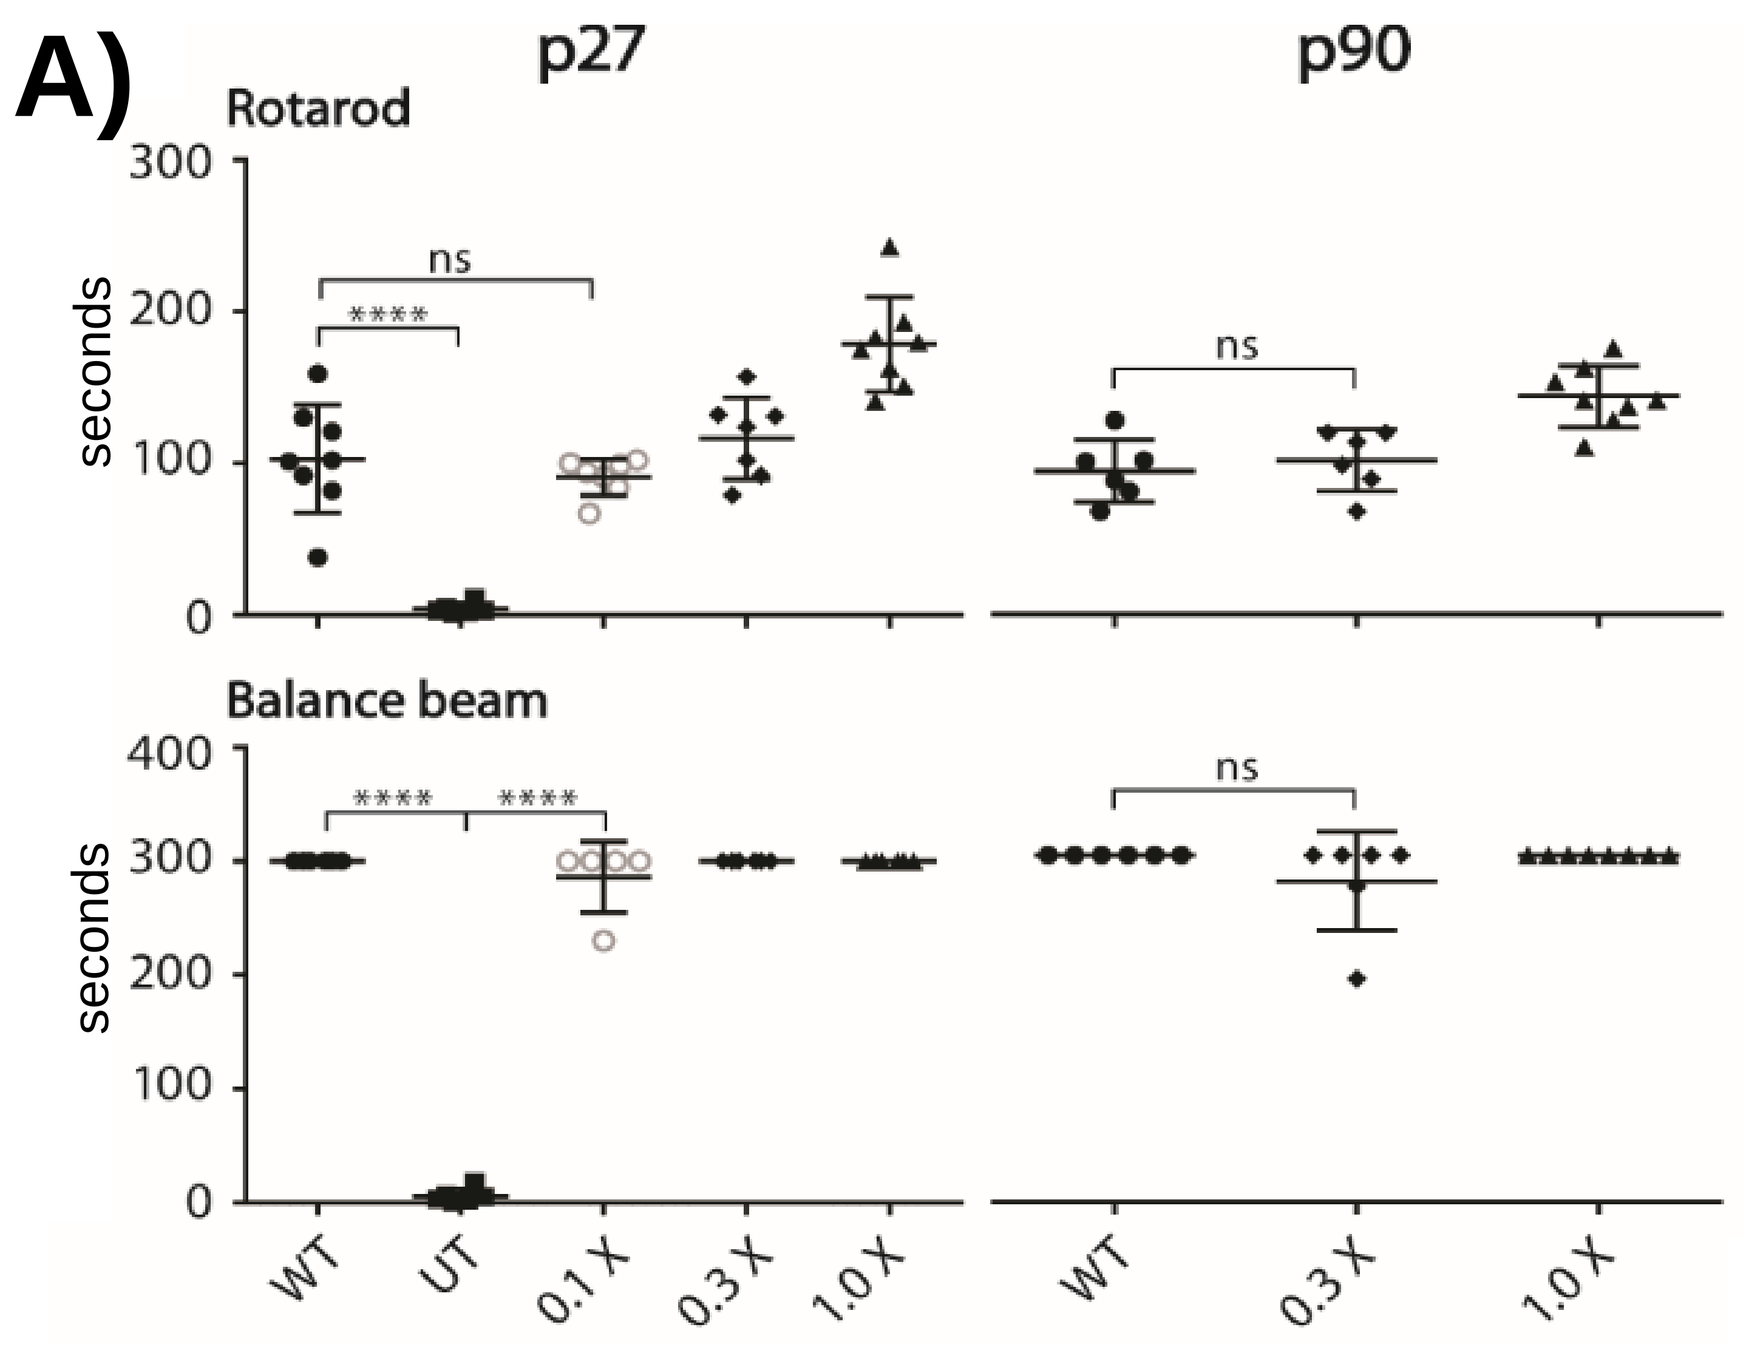
A)
p27
p90
Rotarod
seconds
Balance beam
WT
UT
0.1 X
0.3 X
1.0 X

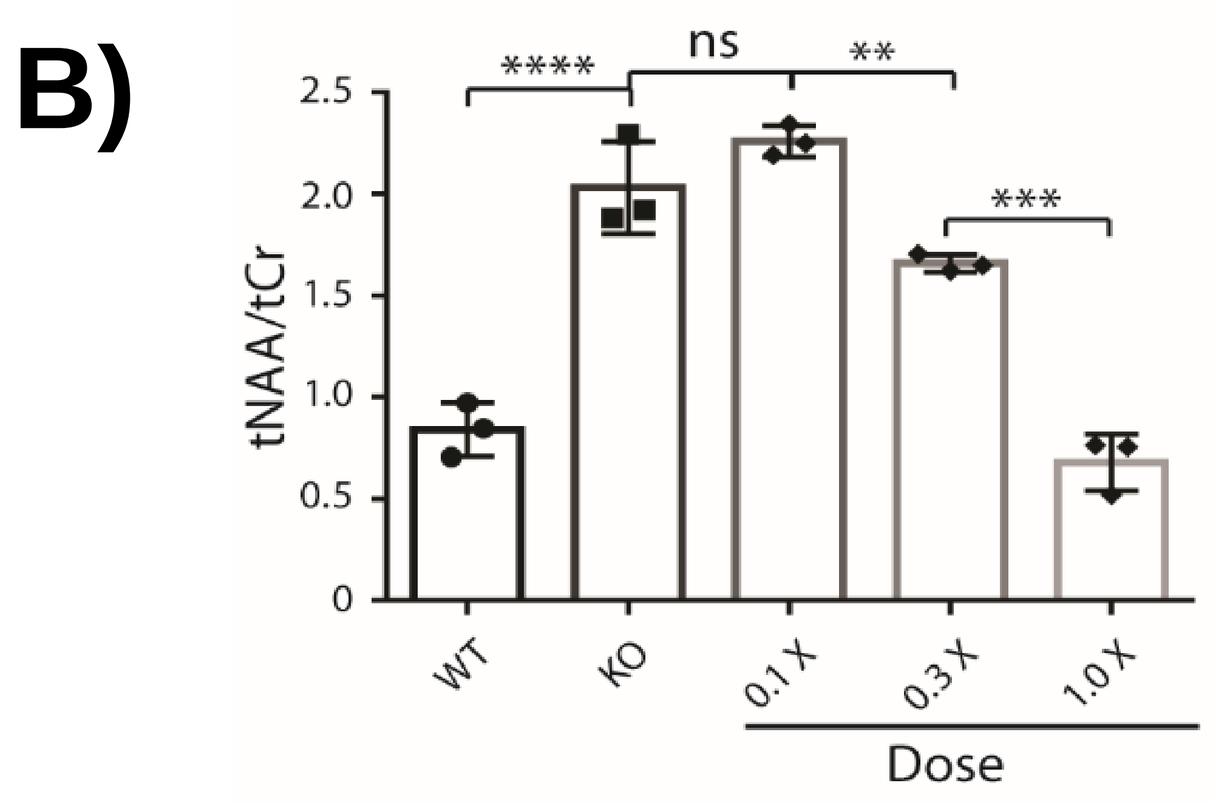
B)
tNAA/tCr
WT
KO
0.1 X
0.3 X
1.0 X
Dose

Figure 1. BBP-812 restores motor function and NAA metabolism in Aspa$^{-/-}$ mice. Mice were treated with PND1 and monitored for motor function 27 or 90 days after birth (A) and had brain NAA levels measured by MRS analysis 25 days after birth.

Data courtsey of Dominic Gessler and Guangping Gao. JCI Insight: 2017 Feb 9;2(3)

Study Overview

Study Design

Group	Treatment	ROA	Dose (vg/kg or brain)	Necropsy Interval (M/F) 3 Weeks (Day 22)	Necropsy Interval (M/F) 8 Weeks (Day 57)
1	BBP-812	IV	1.8×10^{13}	0/0	1/2
2	BBP-812	IV	6.0×10^{13}	0/0	1/2
3	BBP-812	IV	1.8×10^{14}	1/2	1/2
4	BBP-812	ICV	5.0×10^{12}	1/2	1/2
5	BBP-812	IT	5.0×10^{12}	1/2	1/2

F = Female
M = Male
ROA = Route of Administration

- 2-2.5 year old cynomolgus macaques
- AAV9 neutralizing antibody negative at 1:5 dilution
- Biodistribution of vector genomes per diploid genome was assessed by ddPCR and transgene mRNA was assessed by qRT-PCR

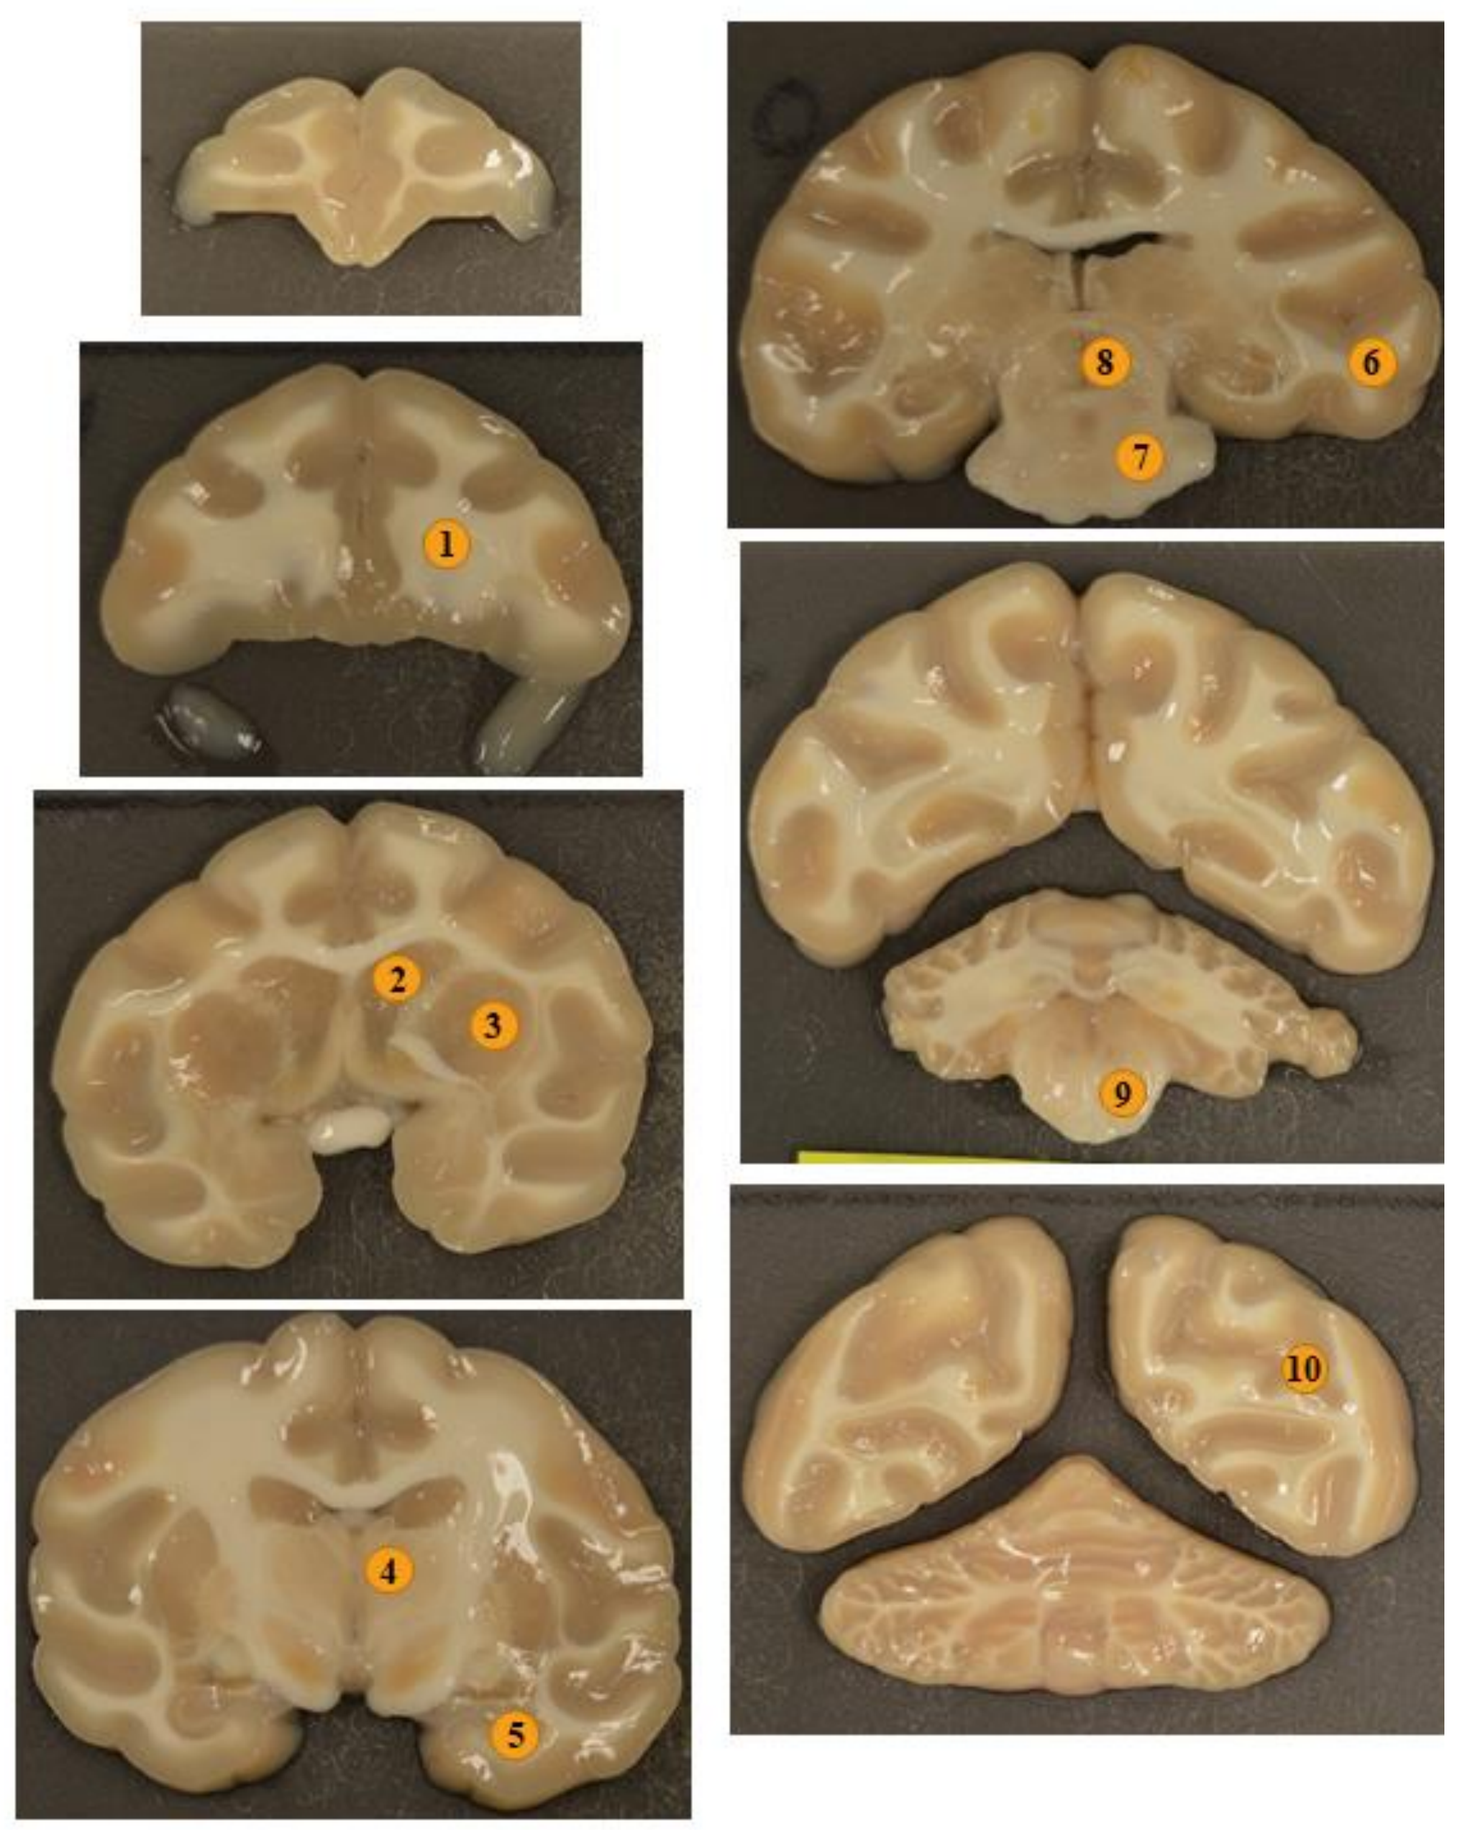

Sample	Tissue
1	Deep frontal cortex
2	Caudate nucleus
3	Putamen
4	Thalamus
5	Hippocampus
6	Temporal cortex
7	Midbrain
8	Pons
9	Medulla
10	Visual cortex

- Animals were routinely monitored for hematology, clinical chemistry, urine chemistry, and CSF chemistry

Results

Broad Central Nervous System Distribution Of BBP-812 Via Multiple Routes Of Administration

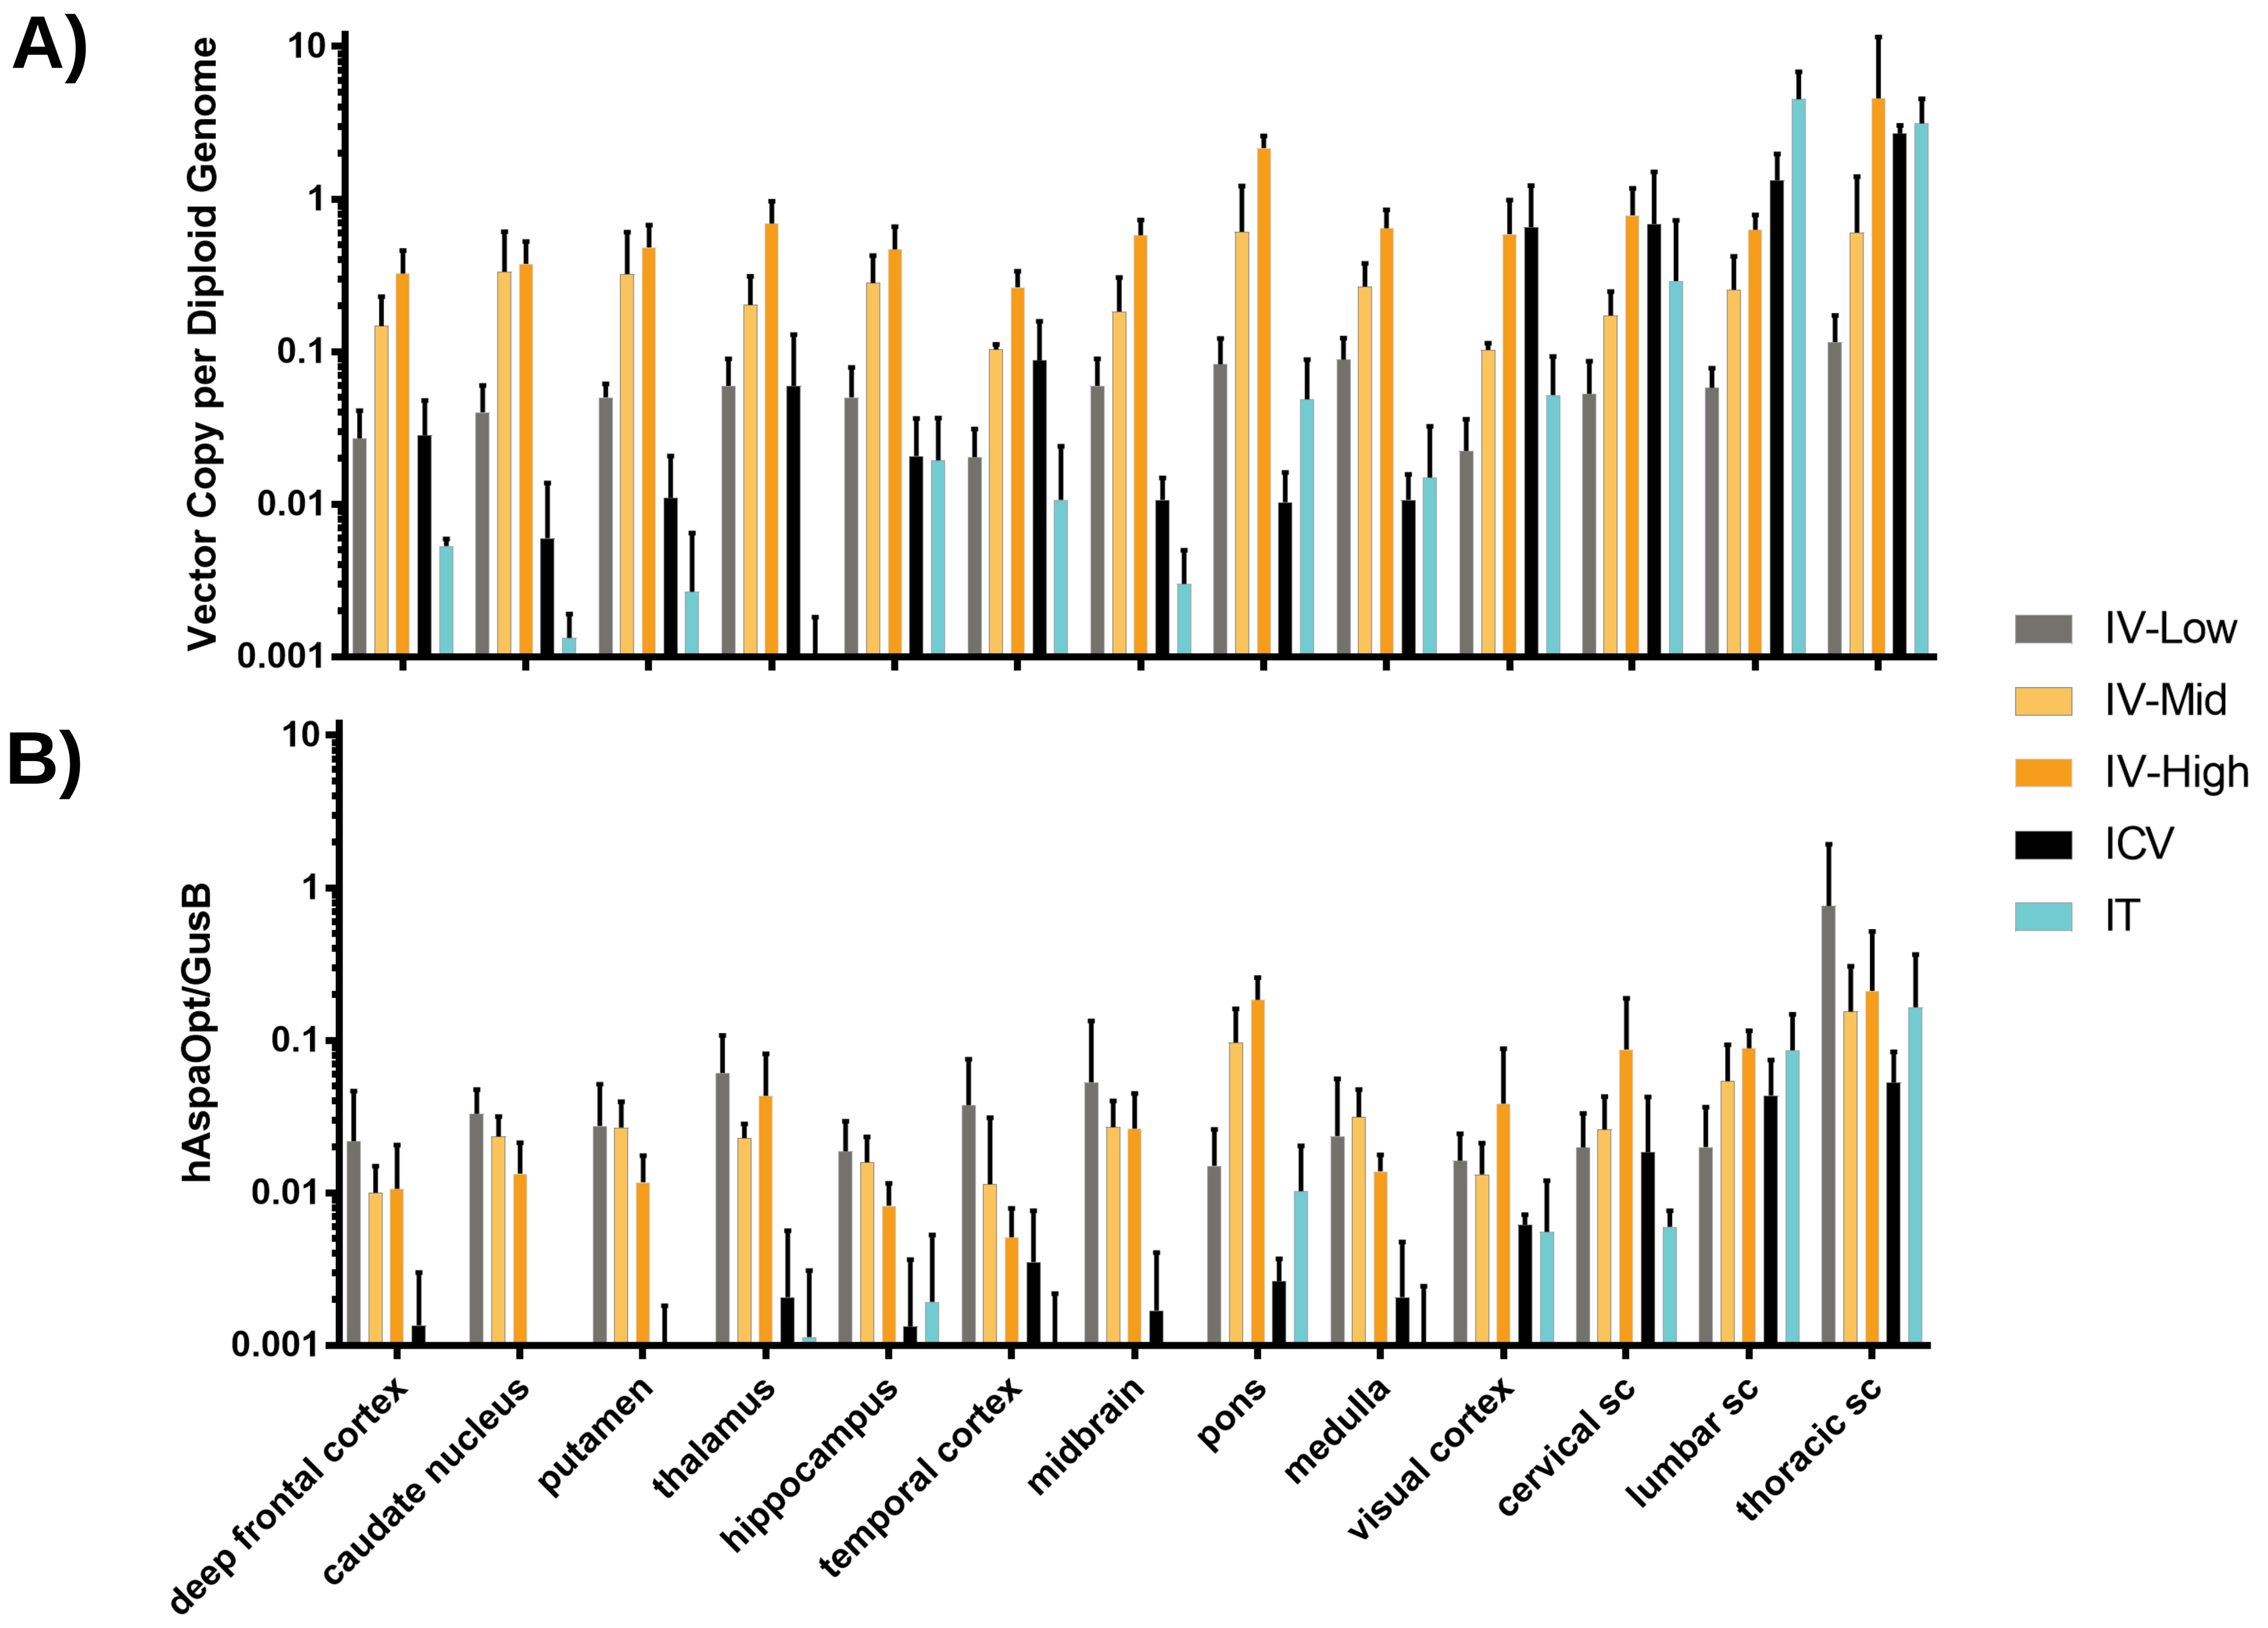
A)
Vector Copy per Diploid Genome
B)
hAspaOpt/GusB
IV-Low
IV-Mid
IV-High
ICV
IT
deep frontal cortex
caudate nucleus
putamen
thalamus
hippocampus
temporal cortex
midbrain
pons
medulla
visual cortex
cervical sc
lumbar sc
thoracic sc

Figure 2. Biodistribution of BBP-812 in the central nervous system. Eight weeks after receiving BBP-812, animals were processed for determination of vector biodistribution in the central nervous system by ddPCR for vector genomes per diploid cell (A) and by qRT-PCR for transgene RNA expression (B).

Transduction Of BBP-812 To Peripheral Target Via Multiple Routes Of Administration

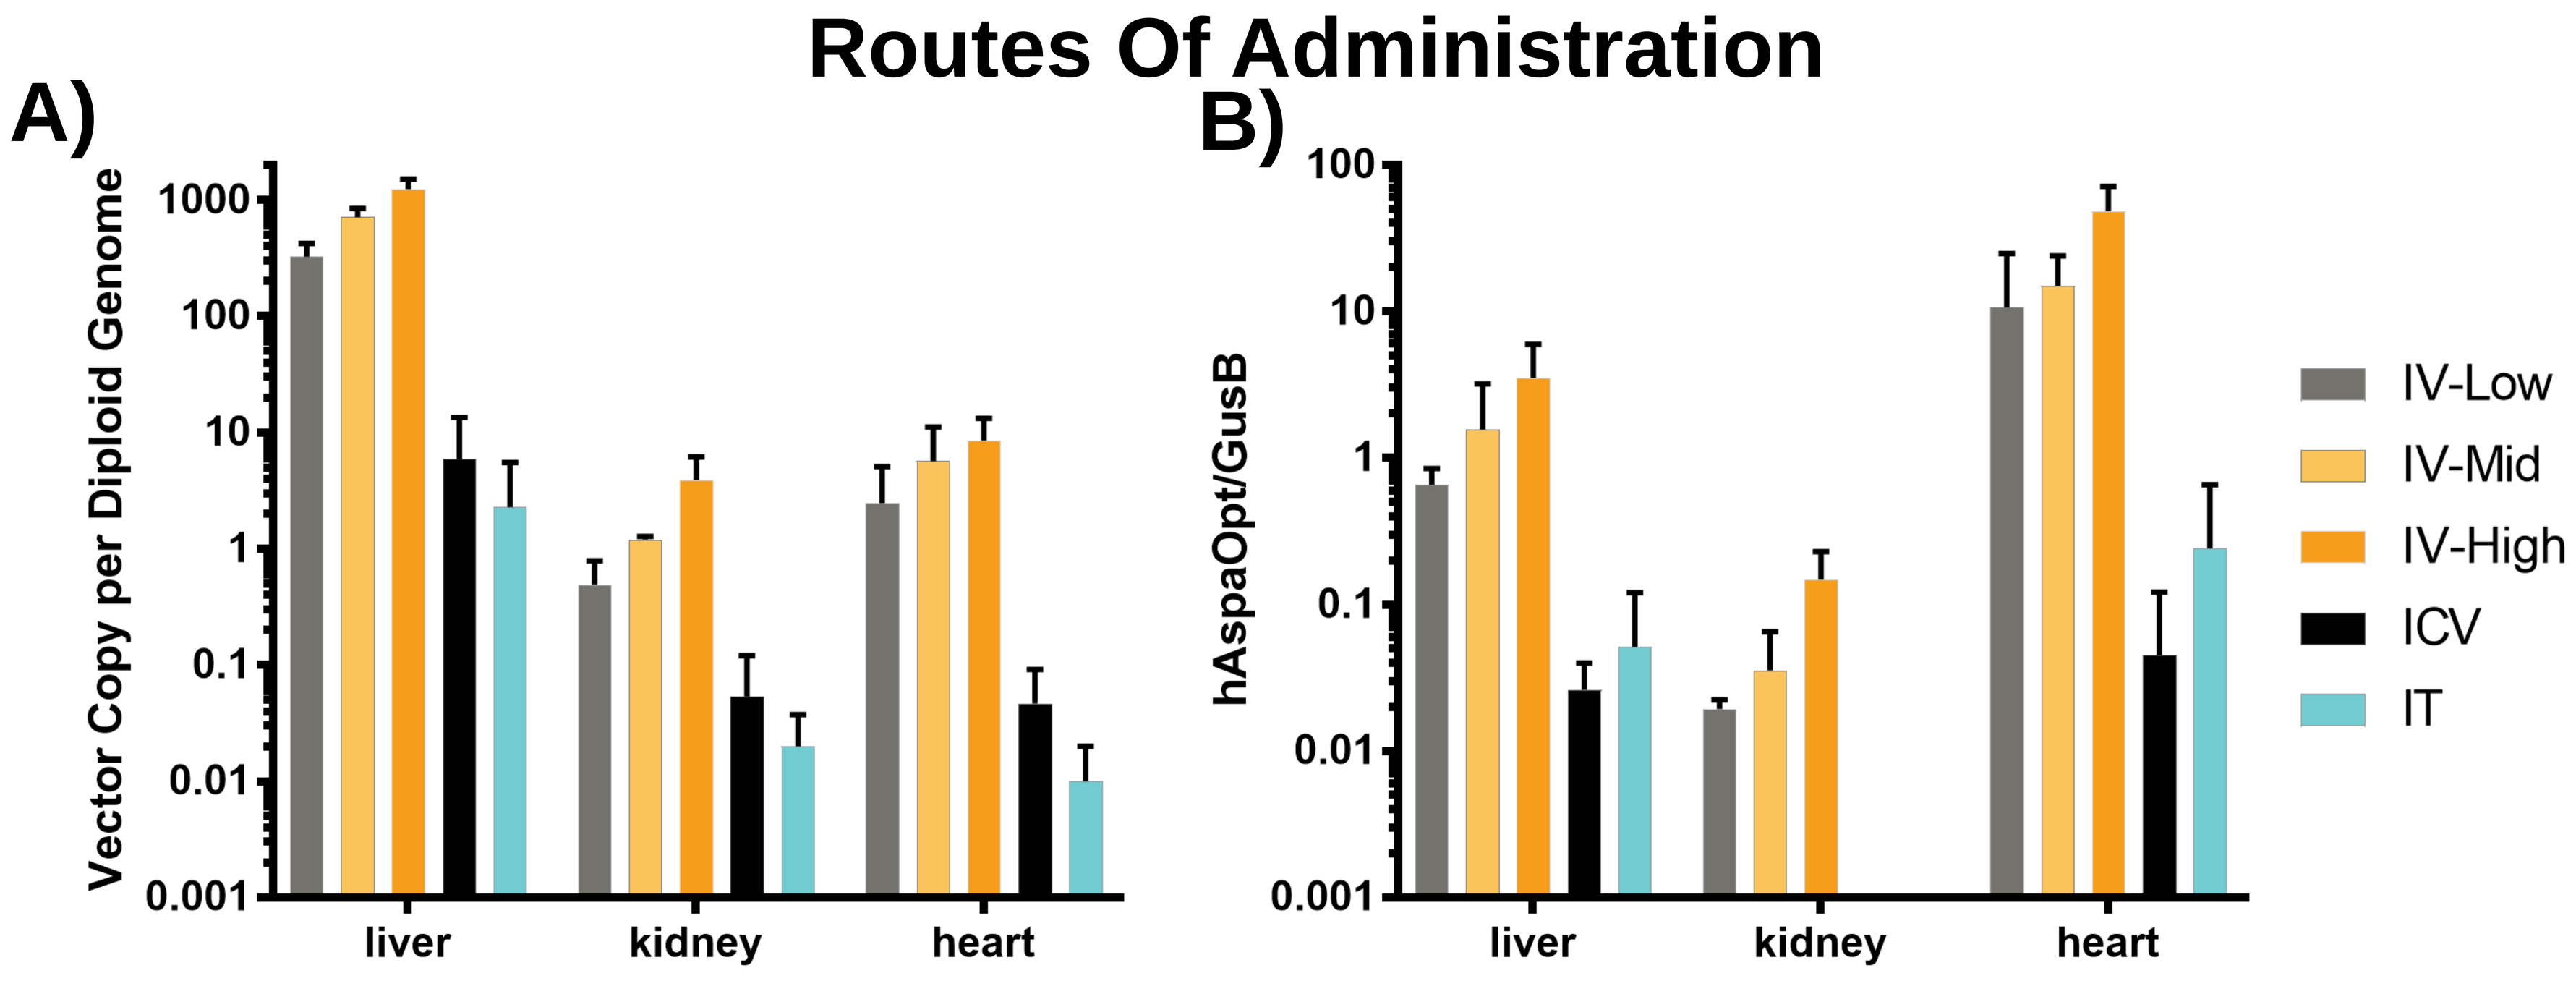
A)
Vector Copy per Diploid Genome
B)
hAspaOpt/GusB
liver
kidney
heart
IV-Low
IV-Mid
IV-High
ICV
IT

Figure 3. Biodistribution of BBP-812 in peripheral tissues. Eight weeks after receiving BBP-812, animals were processed for determination of vector biodistribution in the peripheral tissues by ddPCR for vector genomes per diploid cell (A) and by qRT-PCR for transgene RNA expression (B).

Sustained Expression Of BBP-812 In The Central Nervous System After Intravenous Dosing

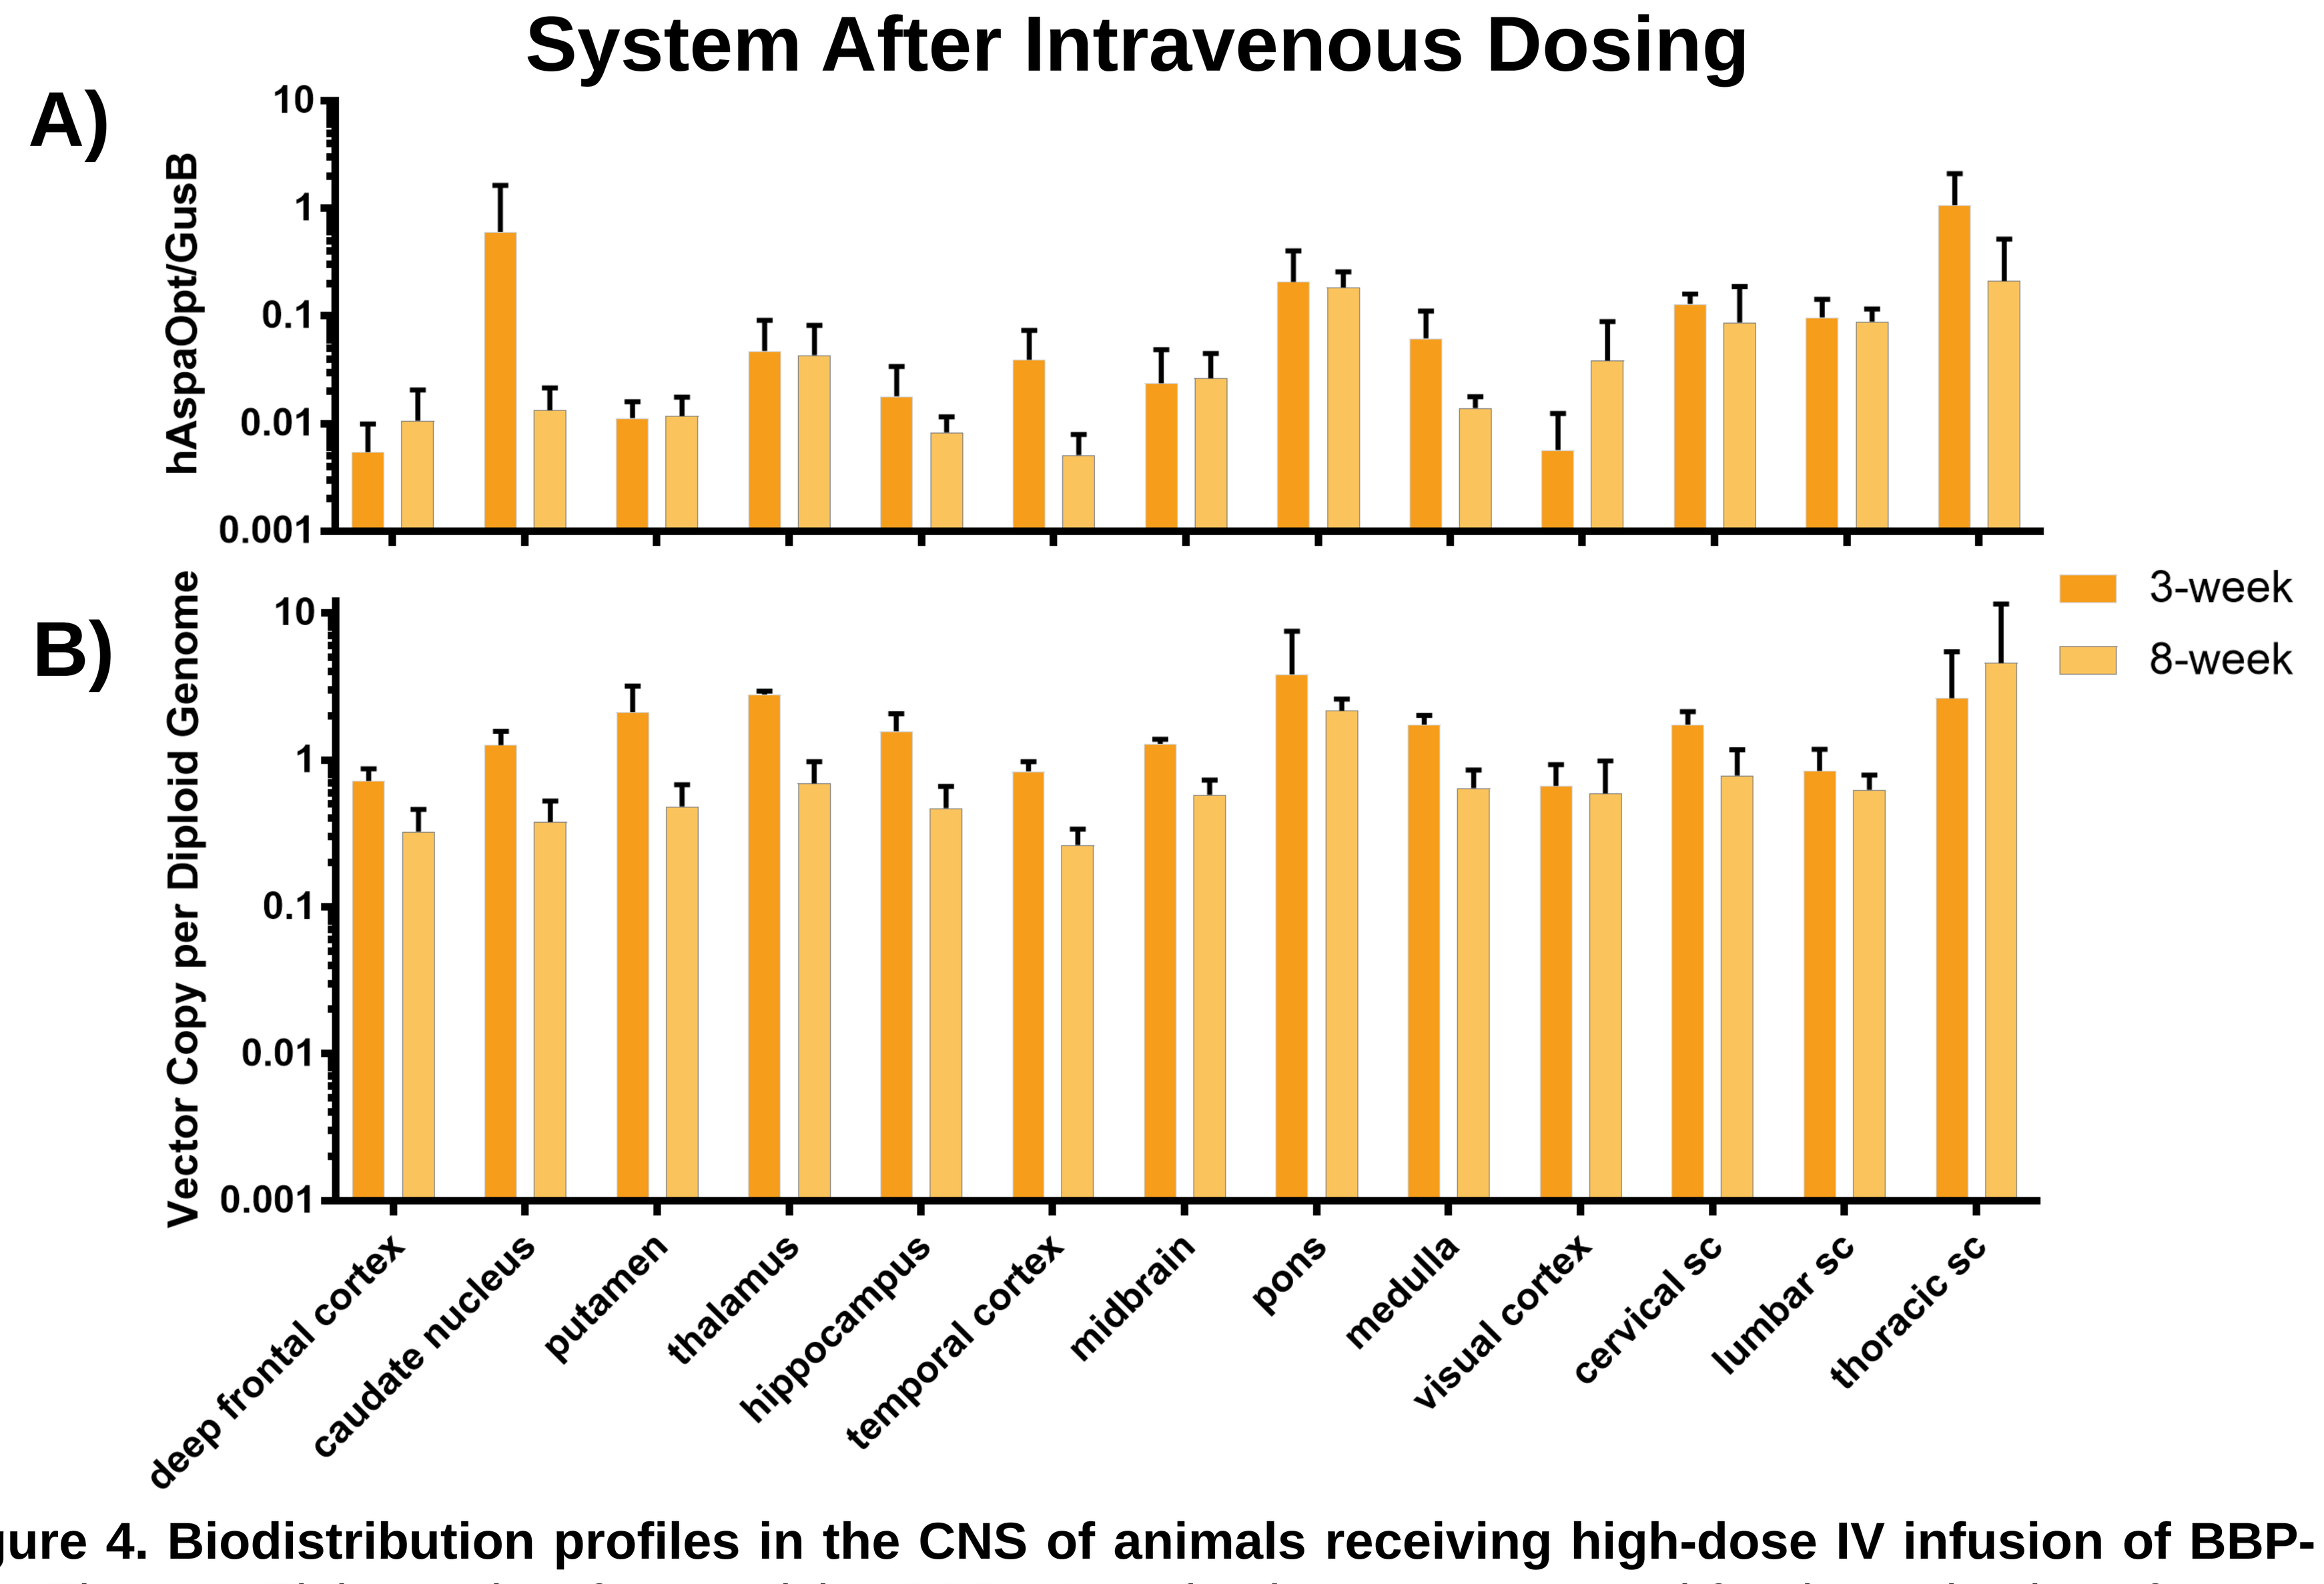
A)
hAspaOpt/GusB
B)
Vector Copy per Diploid Genome
3-week
8-week
deep frontal cortex
caudate nucleus
putamen
thalamus
hippocampus
temporal cortex
midbrain
pons
medulla
visual cortex
cervical sc
lumbar sc
thoracic sc

Figure 4. Biodistribution profiles in the CNS of animals receiving high-dose IV infusion of BBP-812. Three or Eight weeks after receiving BBP-812 animals were processed for determination of vector biodistribution by qRT-PCR for transgene RNA (A) or ddPCR for vector genomes per diploid cell (B)

Conclusions

- No chronic changes in markers of hematology, clinical chemistry, urine chemistry, or CSF chemistry were observed in the study.
- Biodistribution of BBP-812 throughout the CNS was achieved with all ROAs tested with high-dose IV achieving the highest levels of transgene RNA expression.
- Peripheral biodistribution of BBP-812 was achieved by all ROAs.
- **This work supports the continued development of BBP-812 by intravenous infusion for the treatment of Canavan disease.**